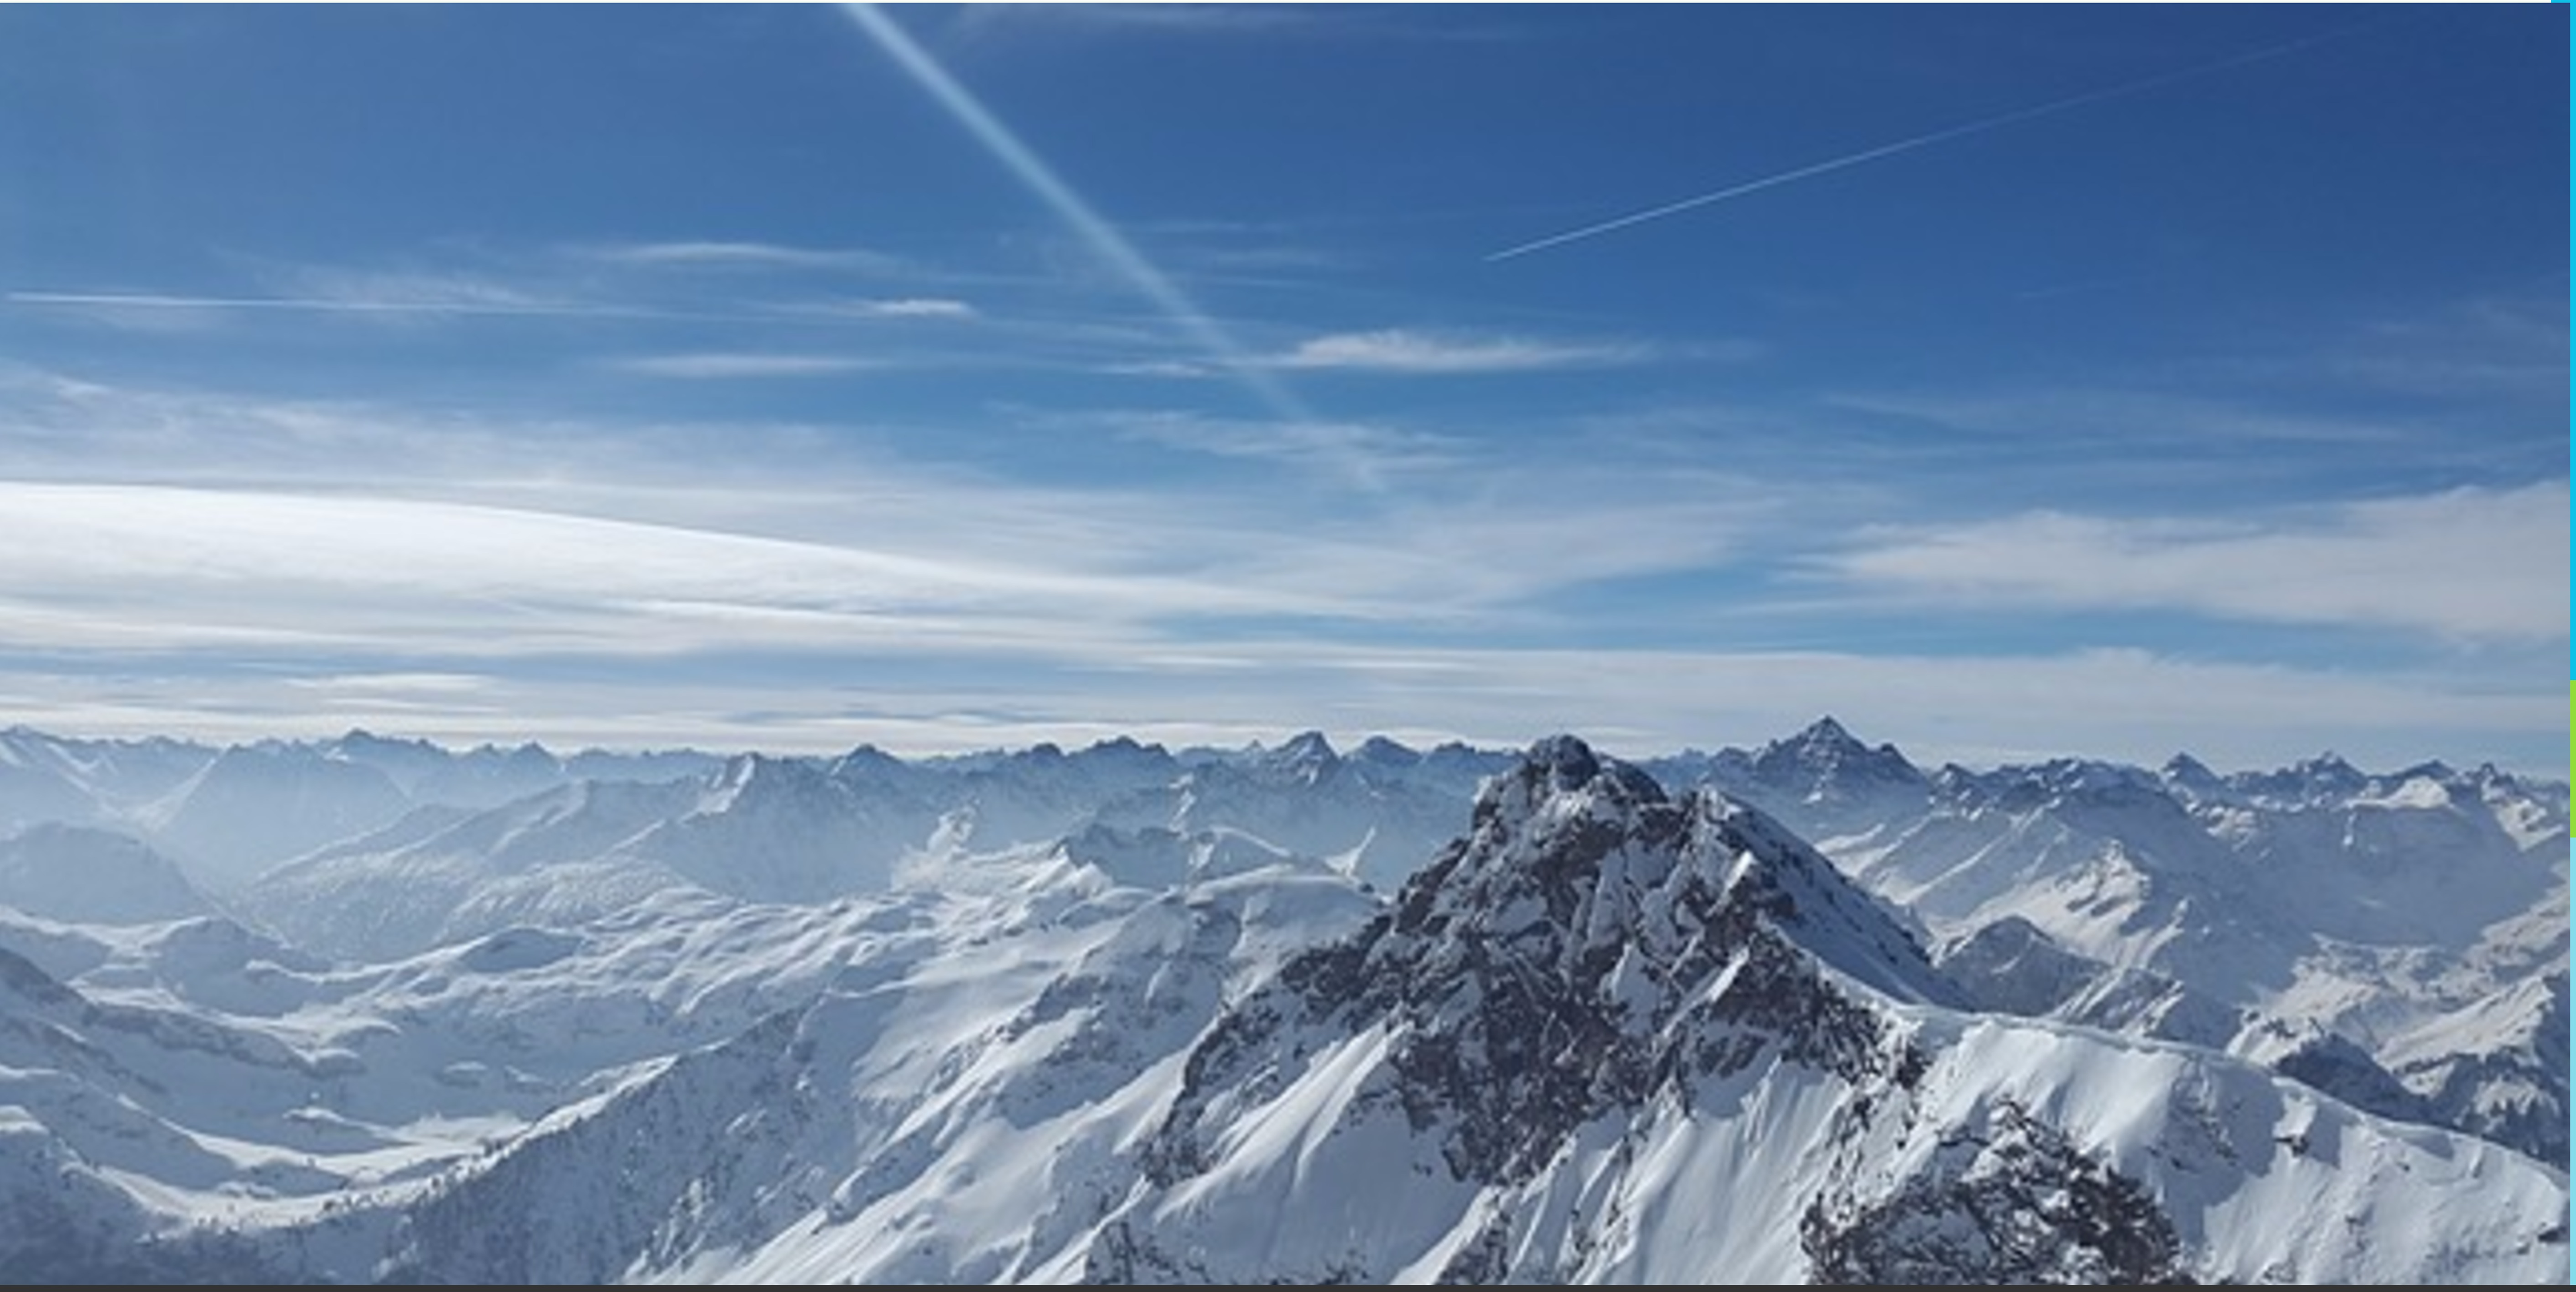

Reaching New Heights: The Glenrose Healthcare Improvement Team

Cyndie Koning, PhD, OT (C), Jennifer Klein, PhD, OT (C), Lesley Pritchard-Wiart, PhD, PT, Laura Mumme, BSc, Alyson Kwok, BScPT, PT, Lisa Froese, BScPT, MPH, Christie Sharun, BSc, MHS, CLSSGB

Context

Why build a Healthcare Improvement Team (HIT)? We needed a structure to facilitate integration of quality improvement activities into practice, so that improving care for patients becomes a part of everything we do at the Glenrose Rehabilitation Hospital. The HIT began in 2014!

What is the purpose of the HIT? The HIT supports healthcare teams to improve patient care by: moving research into practice (knowledge mobilization), engaging in clinical research, and evaluating processes and outcomes.

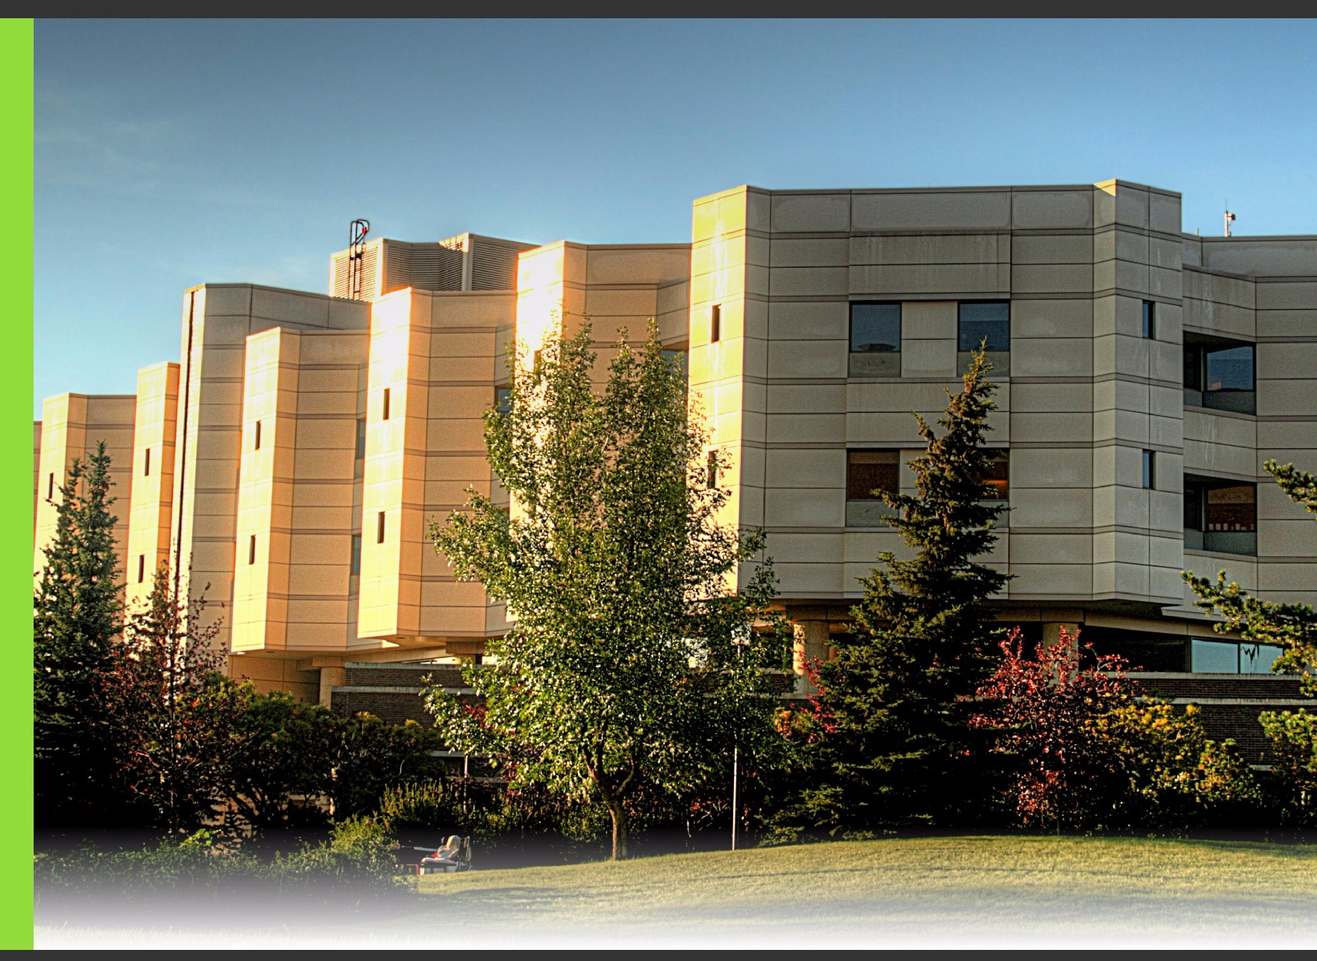

Our journey

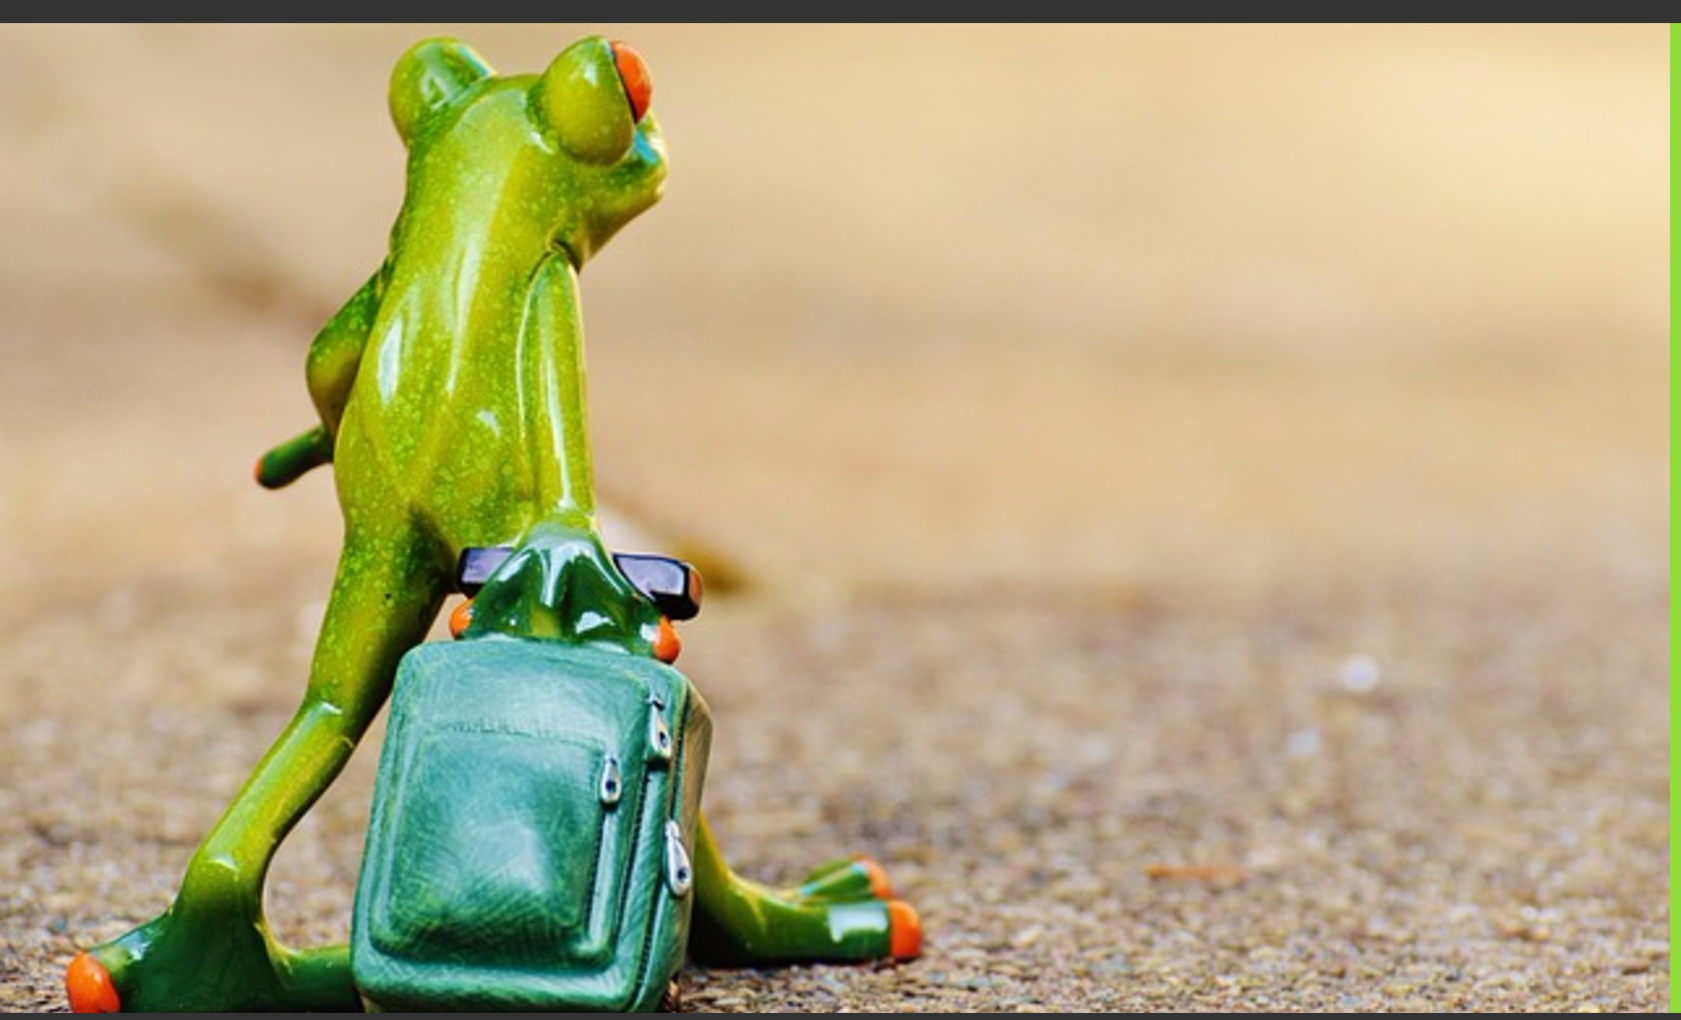

Getting everyone on the same page; identifying partners → Creating clear goals → Building energy to support changes in how we do things → Teaching others what knowledge mobilization and quality improvement means → Listening to patients, clinicians, leaders → Sharing successes, finding opportunities to improve

Impact

45 teams supported by HIT

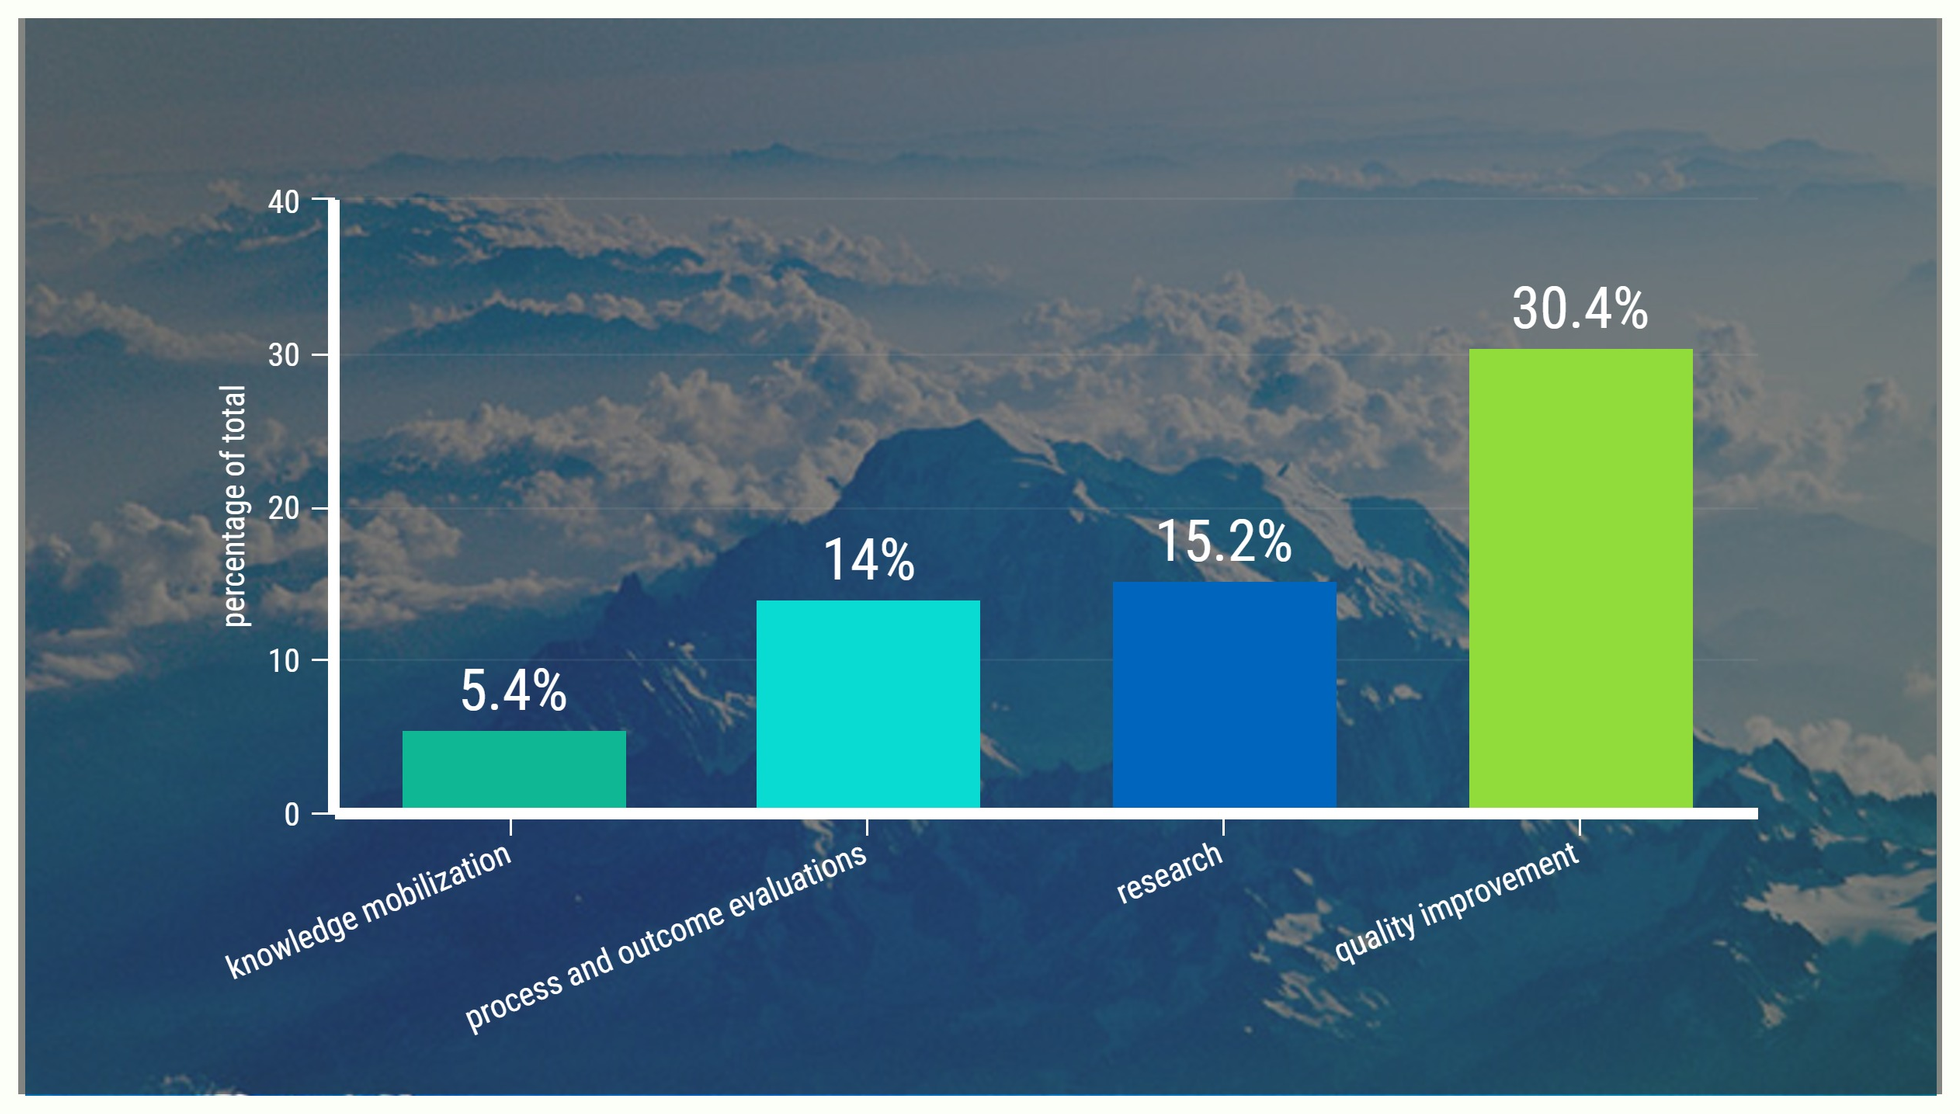
40
30
20
10
0
percentage of total
5.4%
14%
15.2%
30.4%
knowledge mobilization
process and outcome evaluations
research
quality improvement

83 presentations

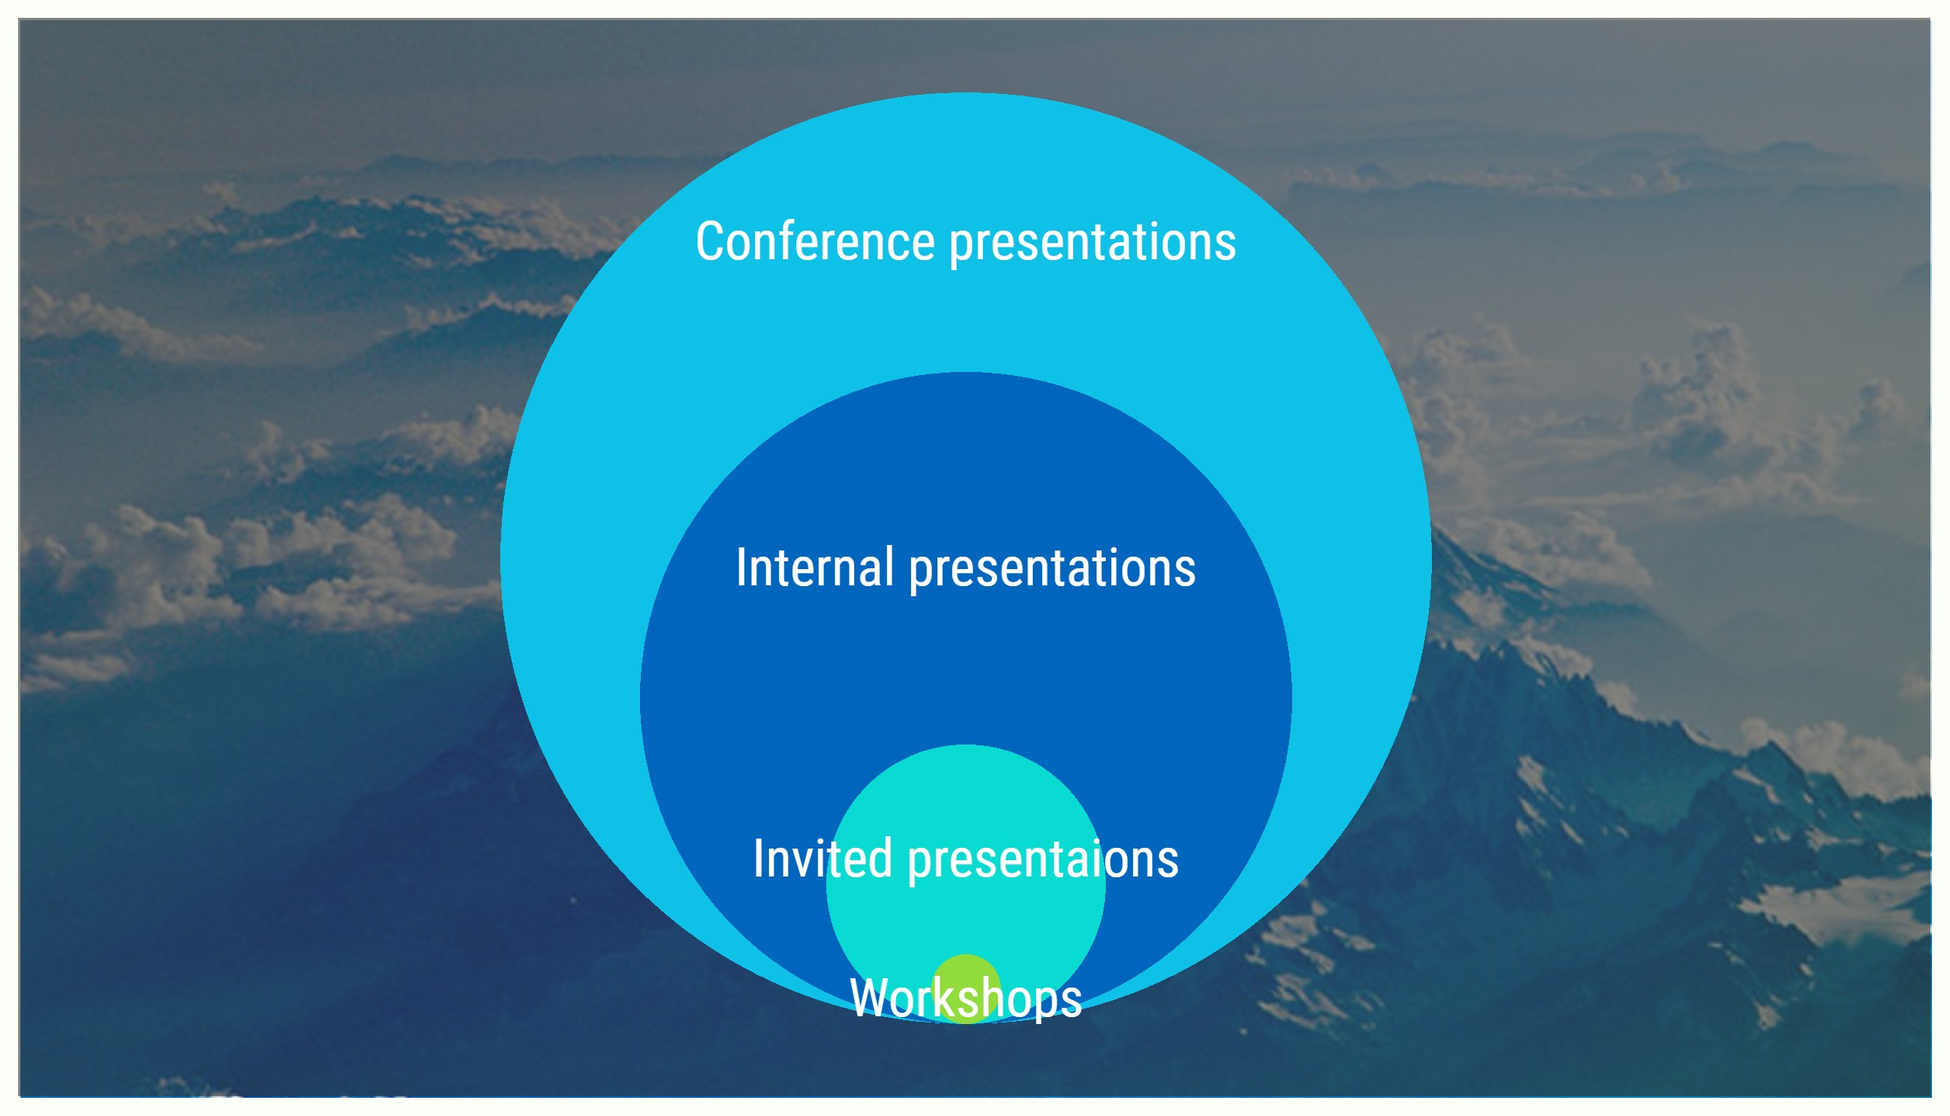
Conference presentations
Internal presentations
Invited presentaions
Workshops

$620,000 in grants

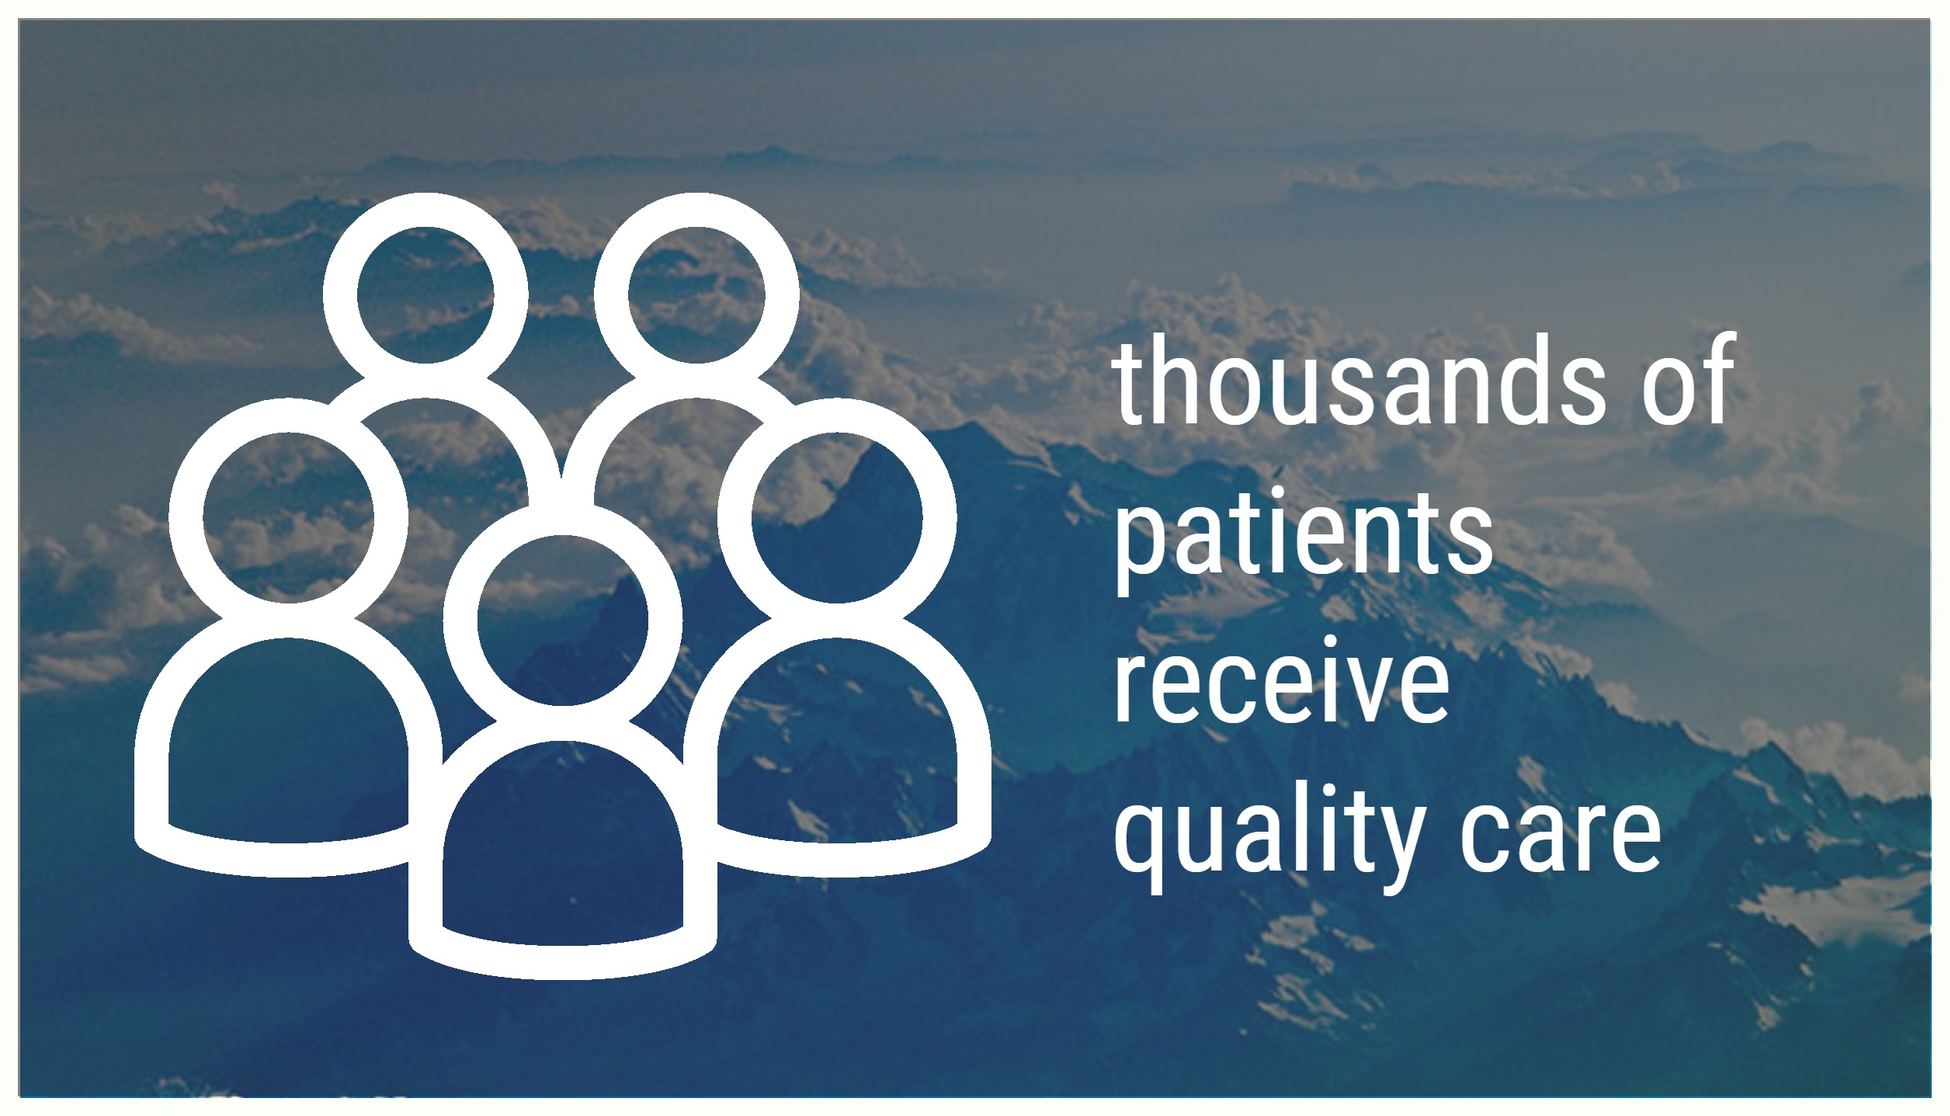
thousands of patients receive quality care

links of projects to Alberta Quality Matrix (past 6 months)

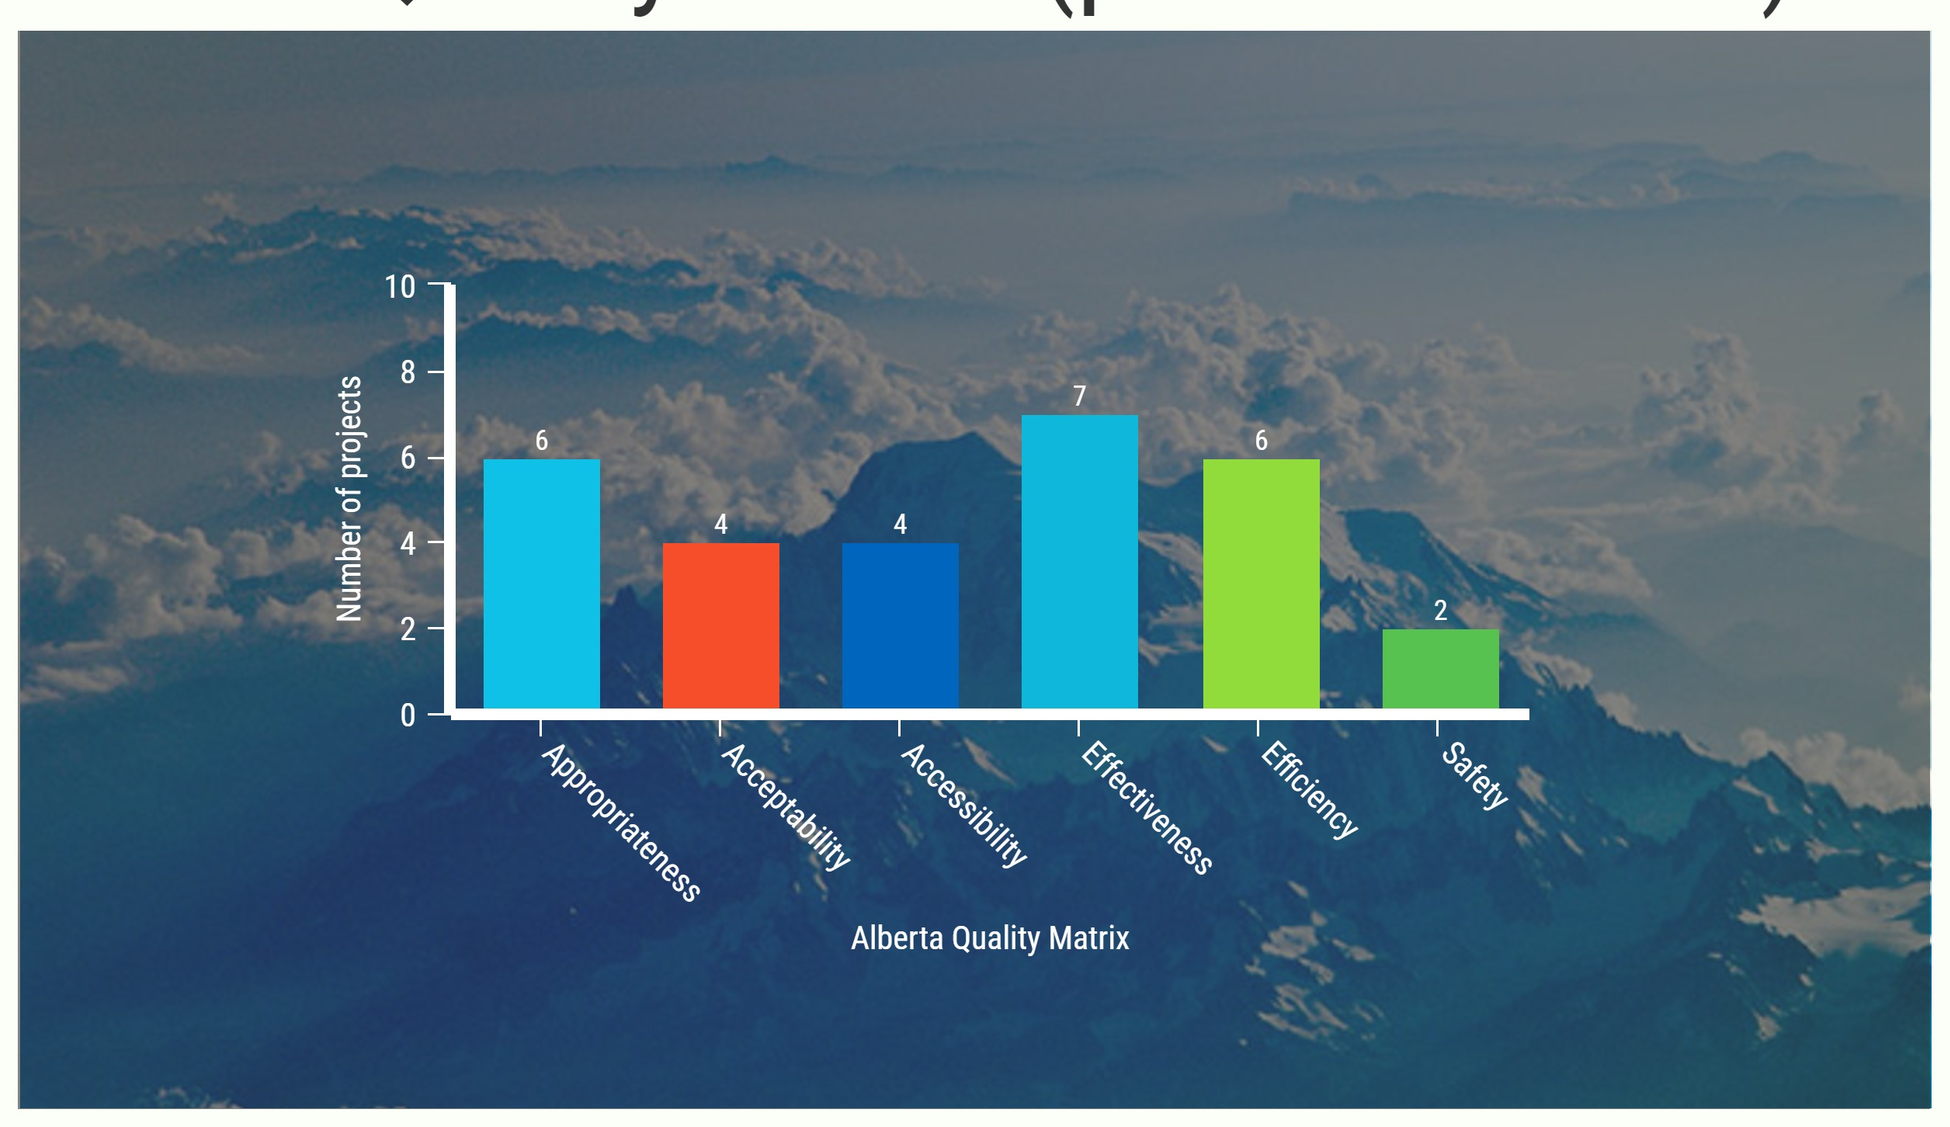
10
8
6
4
2
0
Number of projects
6
4
4
7
6
2
Appropriateness
Acceptability
Accessibility
Effectiveness
Efficiency
Safety
Alberta Quality Matrix

Lessons learned

- Protected time and resources are needed to build capacity and measure impact.
- Within teams, fostering a shared understanding regarding the purpose and key outcomes is crucial.
- Identify and empower a frontline champion. This is key to successfully changing what clinicians do.
- Identify a sponsor: Leader participation helps overcome barriers!
- Make communication plans that target everyone who needs to know more about what your team is doing. Give information in a way that makes sense to them.
- Engage patient advisors early in the process. Build plans for how to keep the work going.
- Communicate evaluation results and measure ongoing success using PDSA cycles. Sustainable change happens when teams take ownership!

Alberta Health Services
Glenrose Rehabilitation Hospital

UNIVERSITY OF ALBERTA